Impact of lung function decline on health-related quality of life in patients with idiopathic pulmonary fibrosis (IPF)

Michael Kreuter,[1] Wibke Stansen,[2] Susanne Stowasser,[2] Nils Schoof[2]

[1]Center for Interstitial and Rare Lung Diseases, Pneumology and Respiratory Care Medicine, Thoraxklinik, University of Heidelberg, Member of the German Center for Lung Research, Germany; [2]Boehringer Ingelheim International GmbH, Ingelheim, Germany.

Introduction

- Idiopathic pulmonary fibrosis (IPF) is a progressive fibrosing interstitial lung disease (ILD) characterized by worsening dyspnea, loss of lung function and impairment in health-related quality of life (HRQL).[1,2]
- Nintedanib is an approved treatment for IPF. In the two placebo-controlled Phase III INPULSIS® trials, nintedanib significantly reduced disease progression by reducing the annual rate of decline in forced vital capacity (FVC), but was not associated with an improvement in HRQL assessed using the St. George's Respiratory Questionnaire (SGRQ).[3]
- Registry data suggest that lower FVC % predicted and decline in FVC % predicted are associated with worse HRQL in patients with IPF.[4,5]

Aim

- To evaluate the impact of decline in lung function measured by FVC on HRQL in patients with IPF in the INPULSIS® trials.

Methods

- In the INPULSIS® trials, patients with IPF diagnosed in the previous 5 years, FVC ≥50% predicted, and a diffusing capacity of the lungs for carbon monoxide 30–79% predicted were randomized 3:2 to receive nintedanib 150 mg bid or placebo for 52 weeks.
- The following patient-reported outcomes (PROs) were used to assess HRQL:
 - SGRQ: a 50-item questionnaire divided into impact, symptoms and activity domains.[6]
 - University of California San Diego Shortness of Breath Questionnaire (UCSD-SOBQ): a 24-item questionnaire assessing the severity and limitations of dyspnea experienced during activities of daily living with varying levels of exertion.[7]
 - Cough and Sputum Assessment Questionnaire (CASA-Q) cough symptoms and cough impact domains: 11 questions assessing the occurrence and impact of cough.[8]
 - EuroQol 5-Dimensional Quality of Life Questionnaire visual analogue scale (EQ-5D VAS), which assesses general health status.[9]
- For this analysis, data from patients treated with nintedanib or placebo were pooled.
- Absolute changes from baseline in PRO scores at week 52 were analyzed in the following subgroups of patients by absolute change from baseline in FVC % predicted at week 52:
 - Improvement or no decline in FVC % predicted (decline ≤0% predicted)
 - Decline in FVC >0 to ≤5% predicted
 - Decline in FVC >5% to ≤10% predicted
 - Decline in FVC >10% predicted.
- Analyses were descriptive.
- Patients with a missing FVC value at week 52 were excluded. Patients with missing values for a PRO at baseline and/or week 52 were excluded from the analysis of that PRO.

Results

- A total of 1061 patients were treated in the INPULSIS® trials; 864 patients provided data for these analyses. Of these:
 - 253 (29.3%) had an improvement or no decline in FVC % predicted at week 52
 - 249 (28.8%) had a decline in FVC >0 to ≤5% predicted at week 52
 - 201 (23.3%) had a decline in FVC >5 to ≤10% predicted at week 52
 - 161 (18.6%) had a decline in FVC >10% predicted at week 52.

Changes in SGRQ scores

- Changes from baseline in SGRQ scores at week 52 were larger (indicating worse HRQL) with increasing decline in FVC % predicted from baseline to week 52 (Figure 1).
- Patients who had a stable or improved FVC at week 52 also showed stability or improvement in SGRQ score.

Figure 1. *Changes from baseline in SGRQ scores at week 52 by changes in FVC % predicted at week 52*

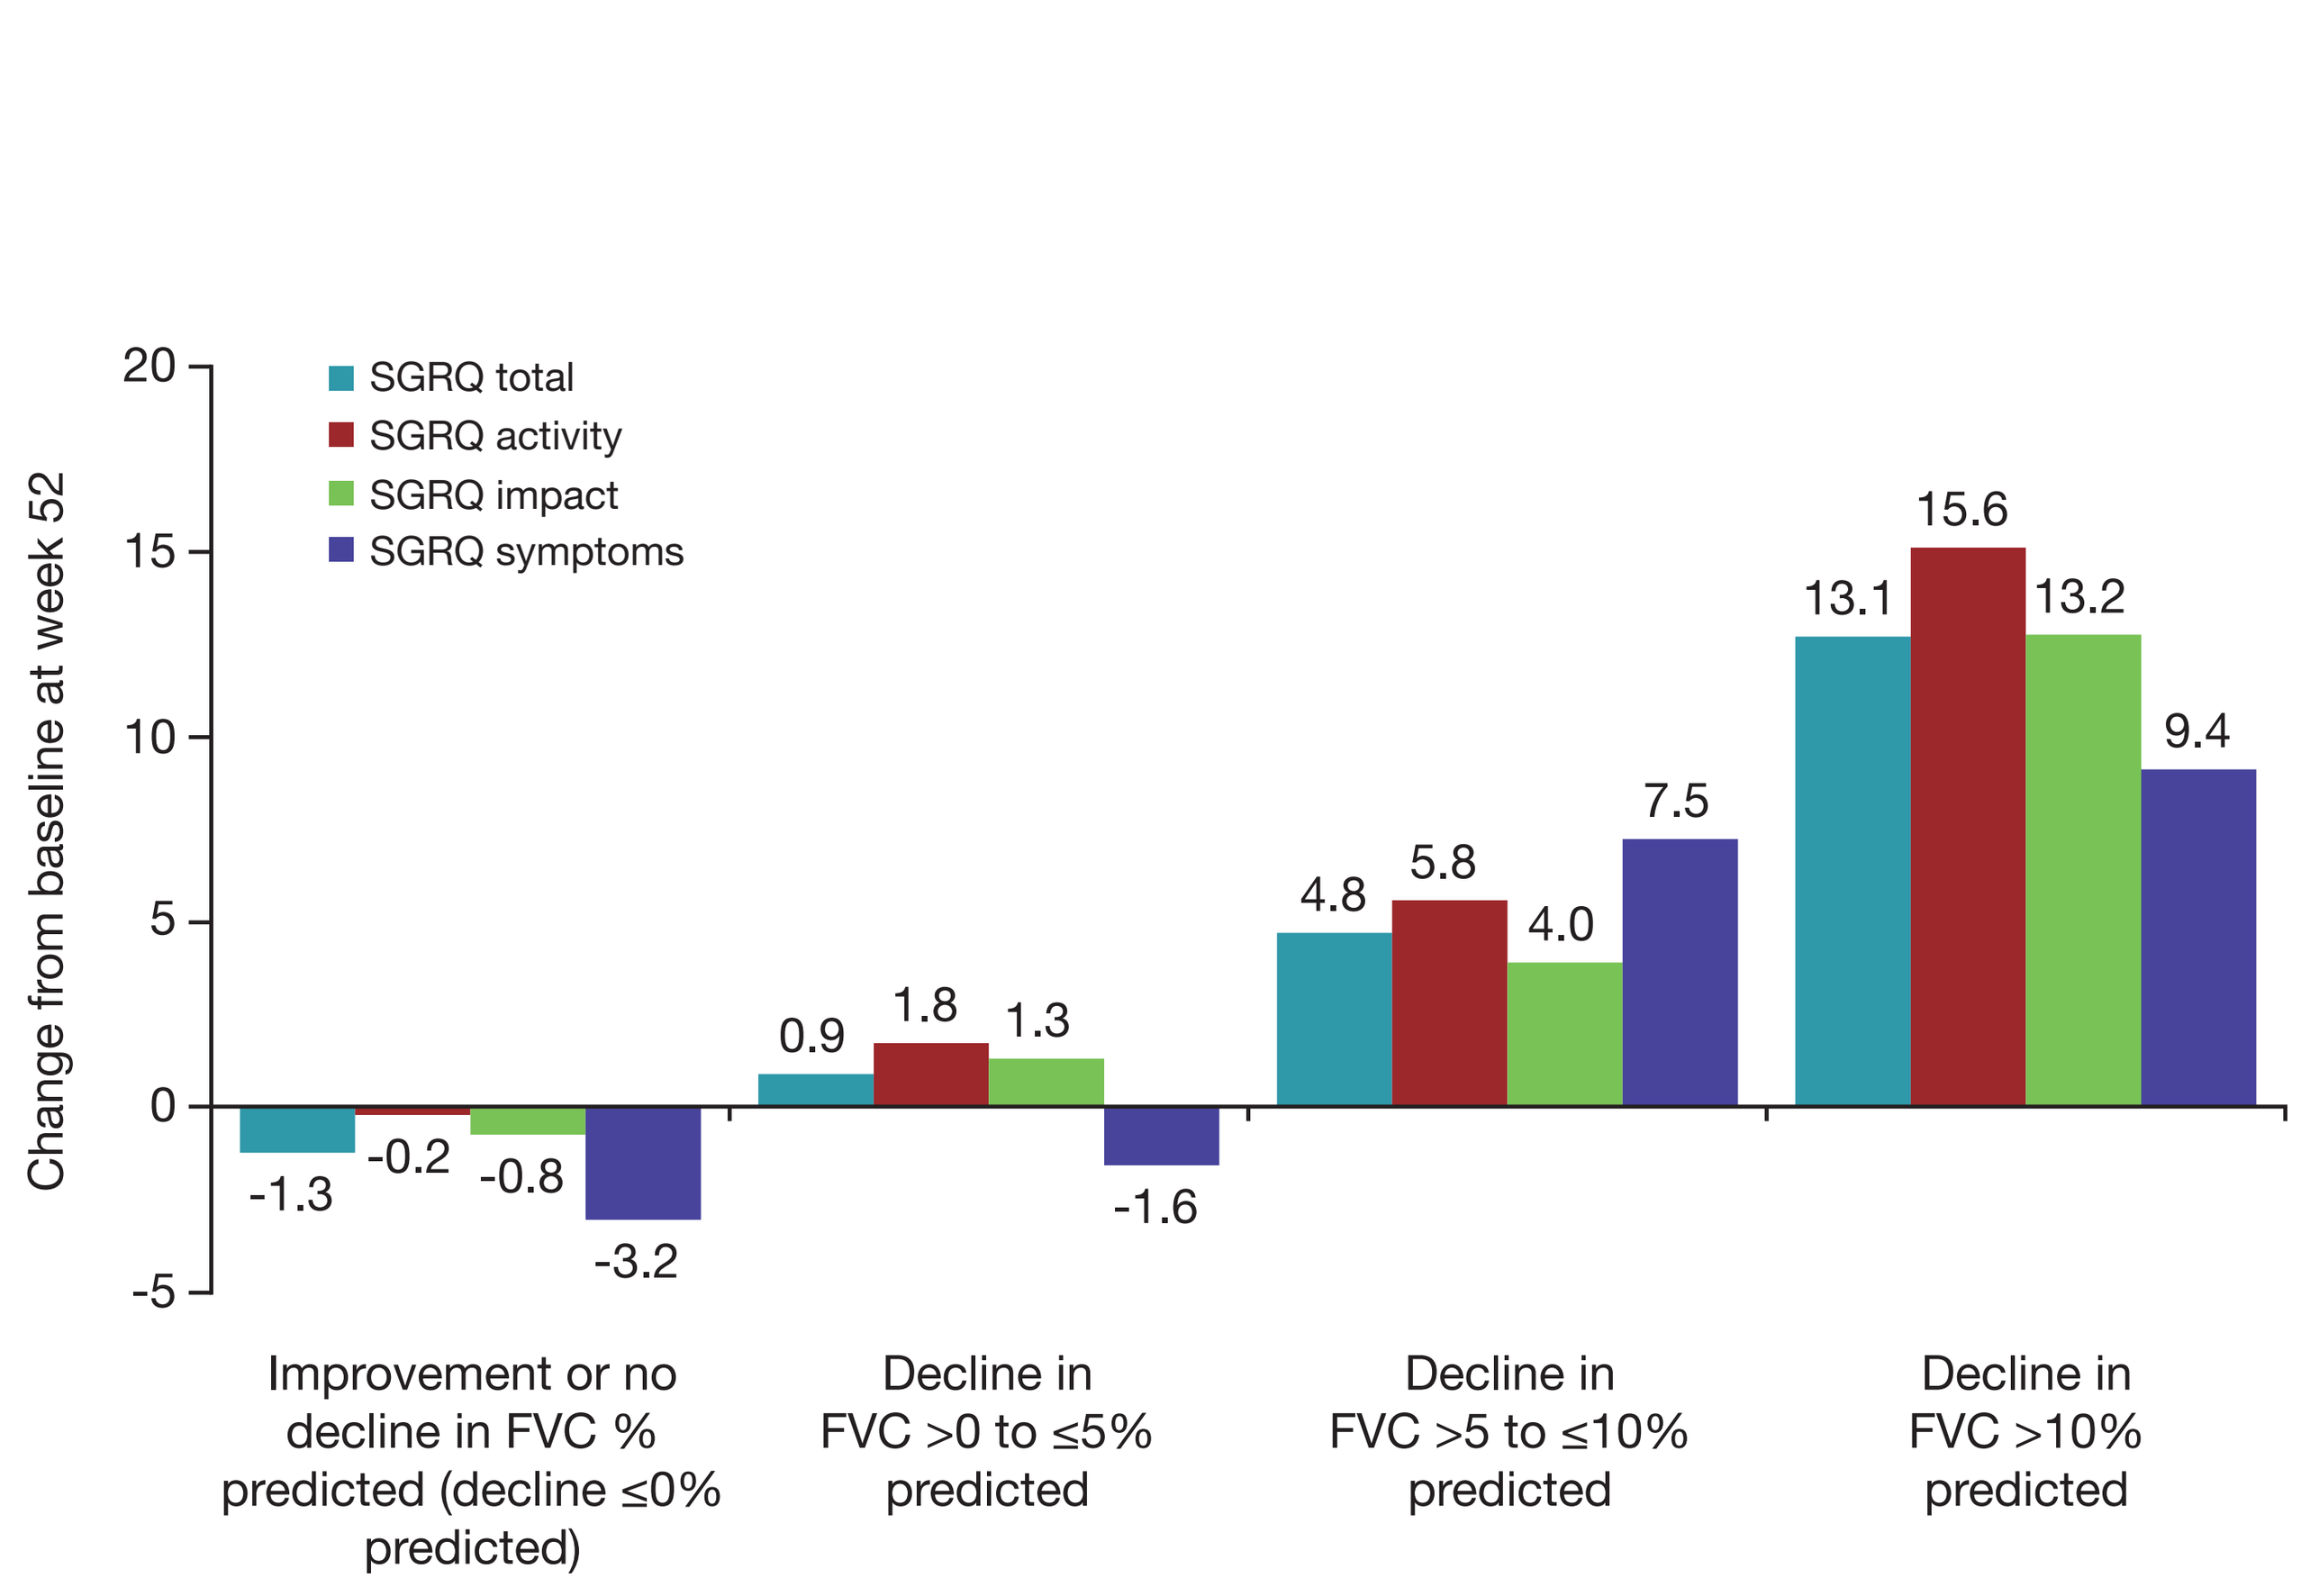

Change from baseline at week 52
20
15
10
5
0
-5
SGRQ total
SGRQ activity
SGRQ impact
SGRQ symptoms
-1.3
-0.2
-0.8
-3.2
0.9
1.8
1.3
-1.6
4.8
5.8
4.0
7.5
13.1
15.6
13.2
9.4
Improvement or no decline in FVC % predicted (decline ≤0% predicted)
Decline in FVC >0 to ≤5% predicted
Decline in FVC >5 to ≤10% predicted
Decline in FVC >10% predicted

Changes in UCSD-SOBQ scores

- The greater the decline in FVC at week 52, the larger the change in UCSD-SOBQ score (Figure 2).

Figure 2. *Changes from baseline in UCSD-SOBQ scores at week 52 by changes in FVC % predicted at week 52*

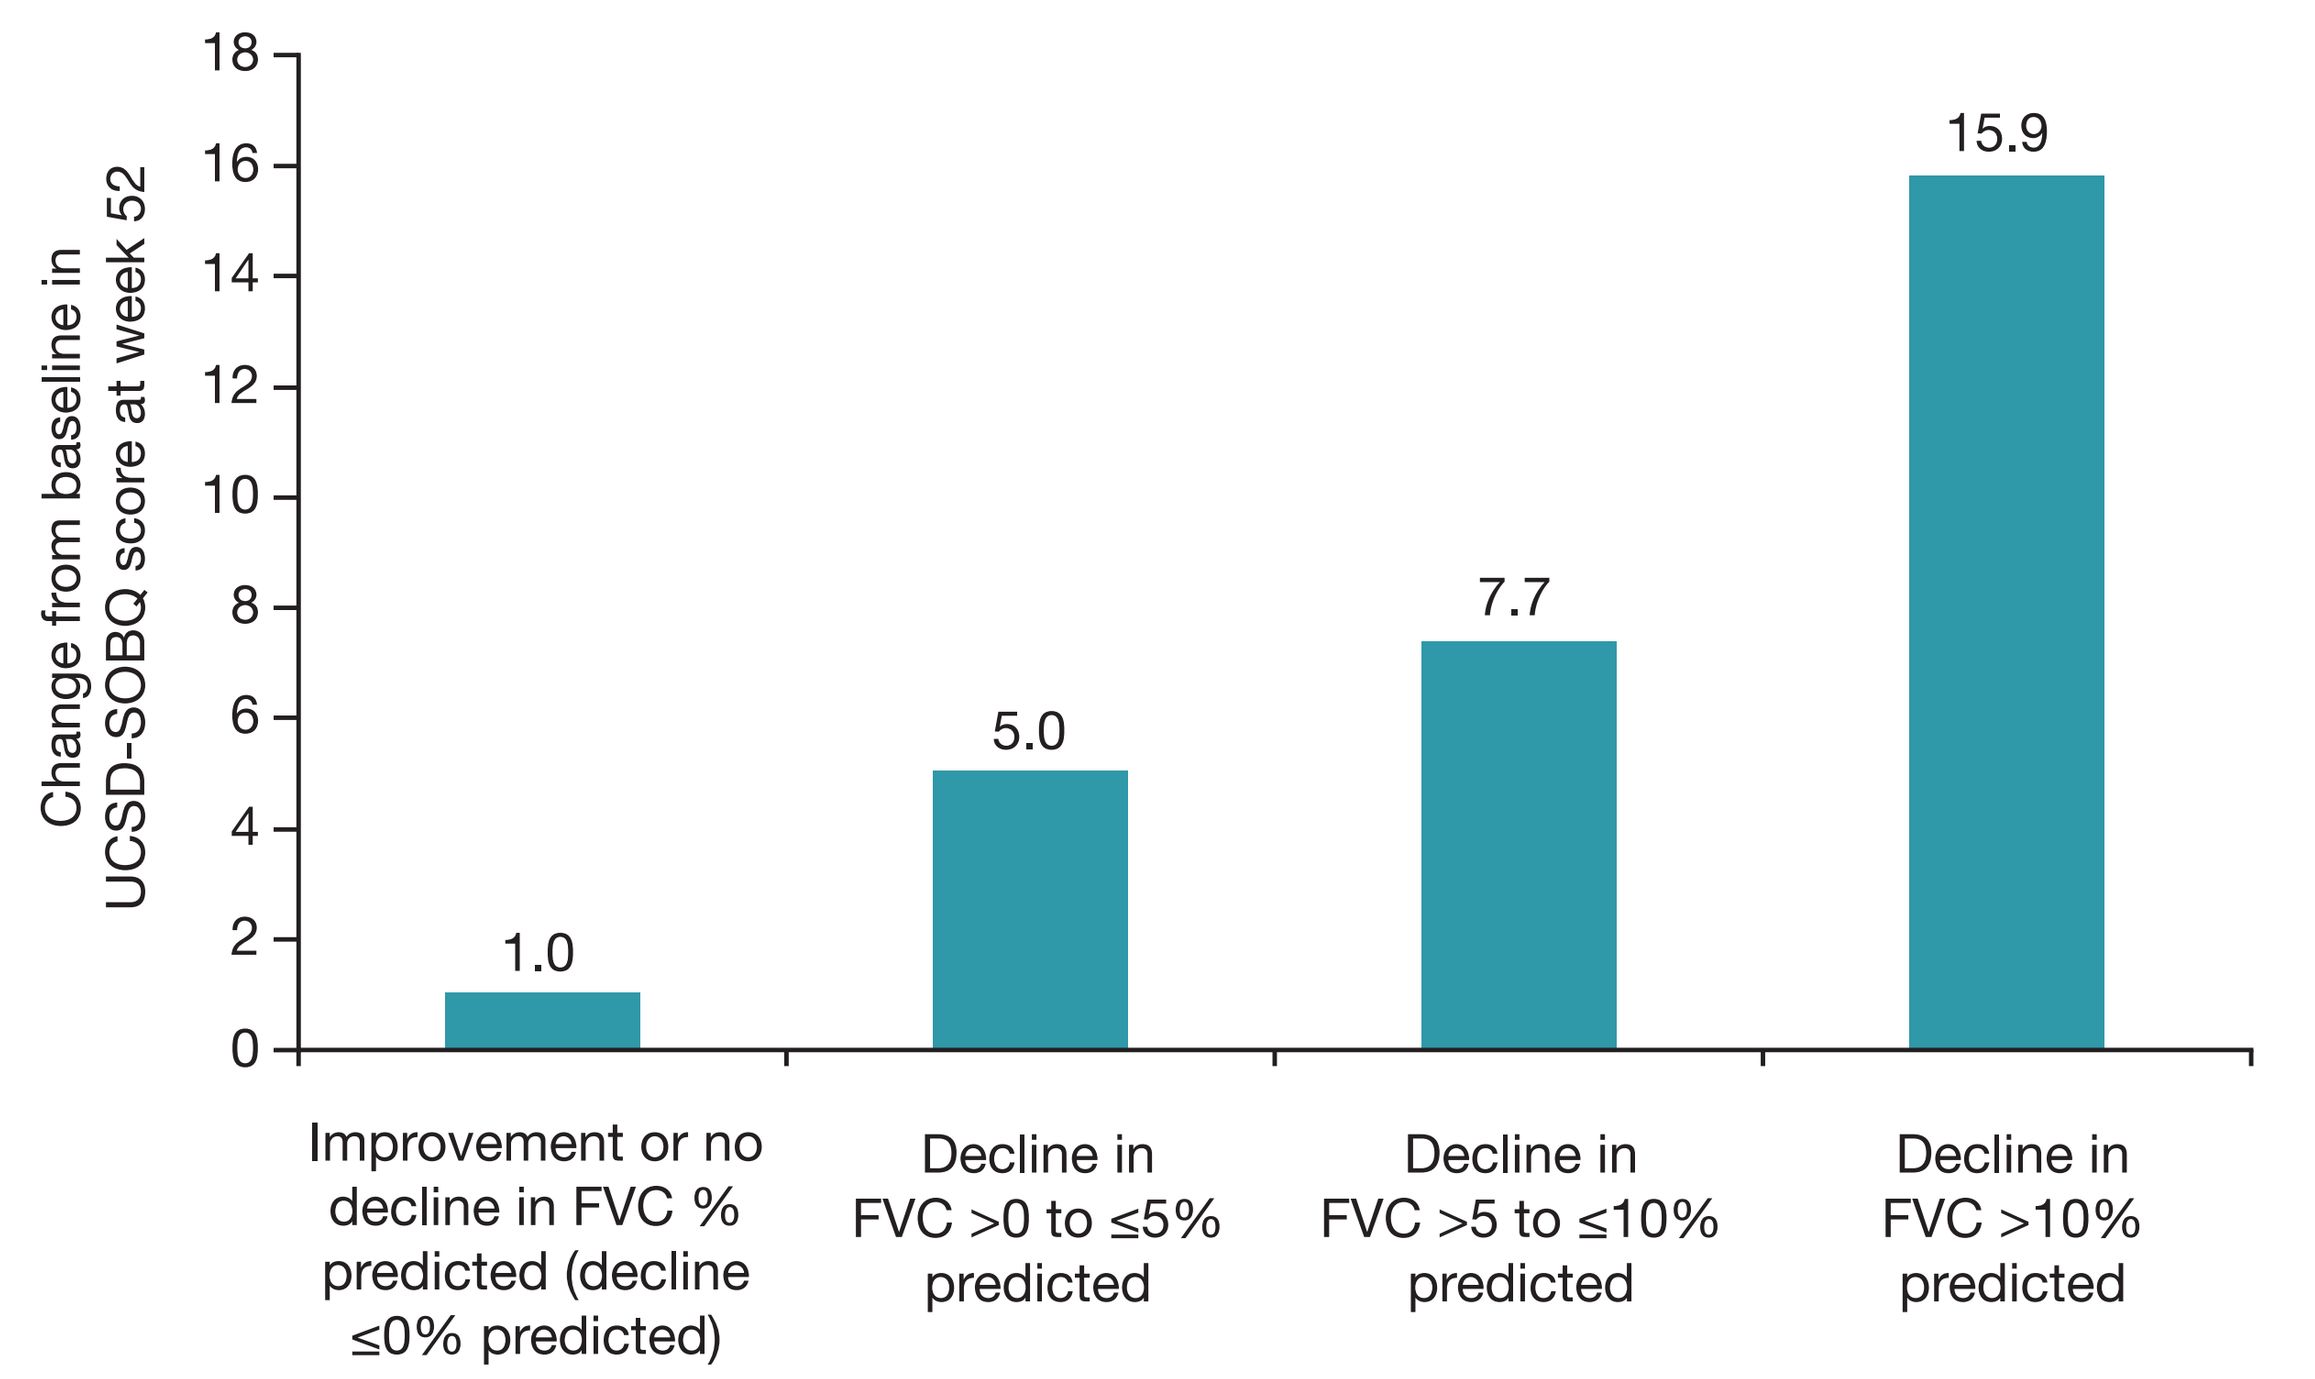

Change from baseline in UCSD-SOBQ score at week 52
18
16
14
12
10
8
6
4
2
0
1.0
5.0
7.7
15.9
Improvement or no decline in FVC % predicted (decline ≤0% predicted)
Decline in FVC >0 to ≤5% predicted
Decline in FVC >5 to ≤10% predicted
Decline in FVC >10% predicted

Changes in CASA-Q cough symptoms and impact scores

- Changes in CASA-Q cough symptoms and impact scores were larger (worse) in patients with a greater decline in FVC at week 52, with greater changes in the cough impact domain than the cough symptoms domain (Figure 3).
- Patients who had a stable or improved FVC at week 52 also showed stability or improvement in the CASA-Q cough symptoms and impact scores.

Figure 3. *Changes from baseline in CASA-Q cough and impact scores at week 52 by changes in FVC % predicted at week 52*

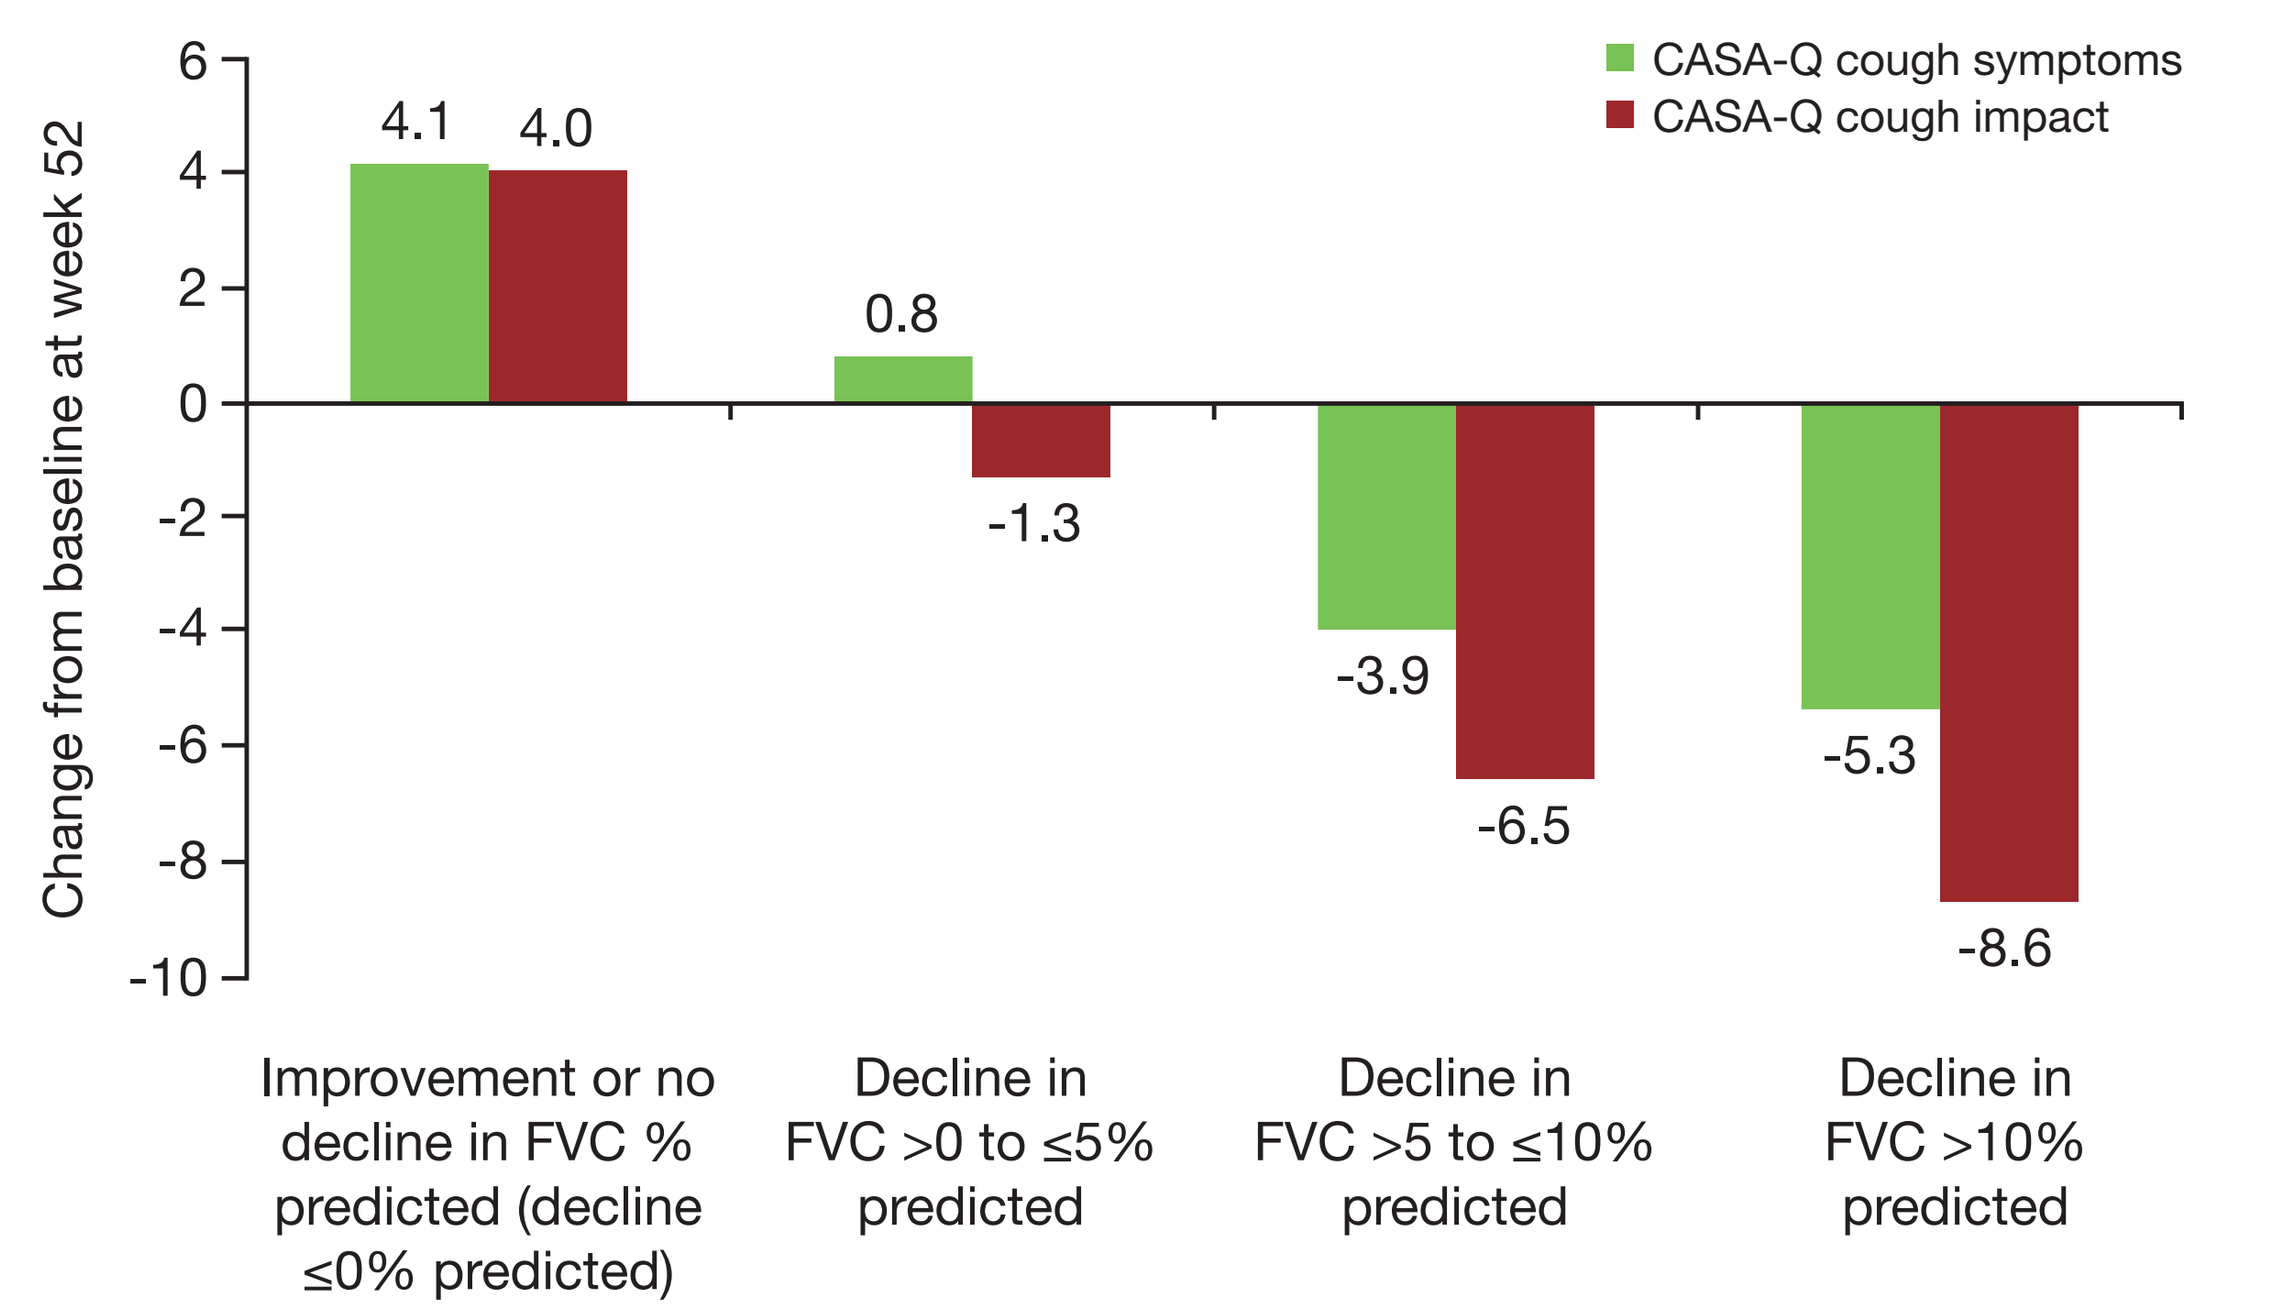

Change from baseline at week 52
6
4
2
0
-2
-4
-6
-8
-10
CASA-Q cough symptoms
CASA-Q cough impact
4.1
4.0
0.8
-1.3
-3.9
-6.5
-5.3
-8.6
Improvement or no decline in FVC % predicted (decline ≤0% predicted)
Decline in FVC >0 to ≤5% predicted
Decline in FVC >5 to ≤10% predicted
Decline in FVC >10% predicted

Changes in EQ-5D VAS score

- Change in EQ-5D VAS score was larger (indicating worse HRQL) in patients with a greater decline in FVC at week 52 (Figure 4).
- Patients who had a stable or improved FVC at week 52 also showed stability or improvement in EQ-5D VAS score.

Figure 4. *Changes from baseline in EQ-5D VAS score at week 52 by changes in FVC % predicted at week 52*

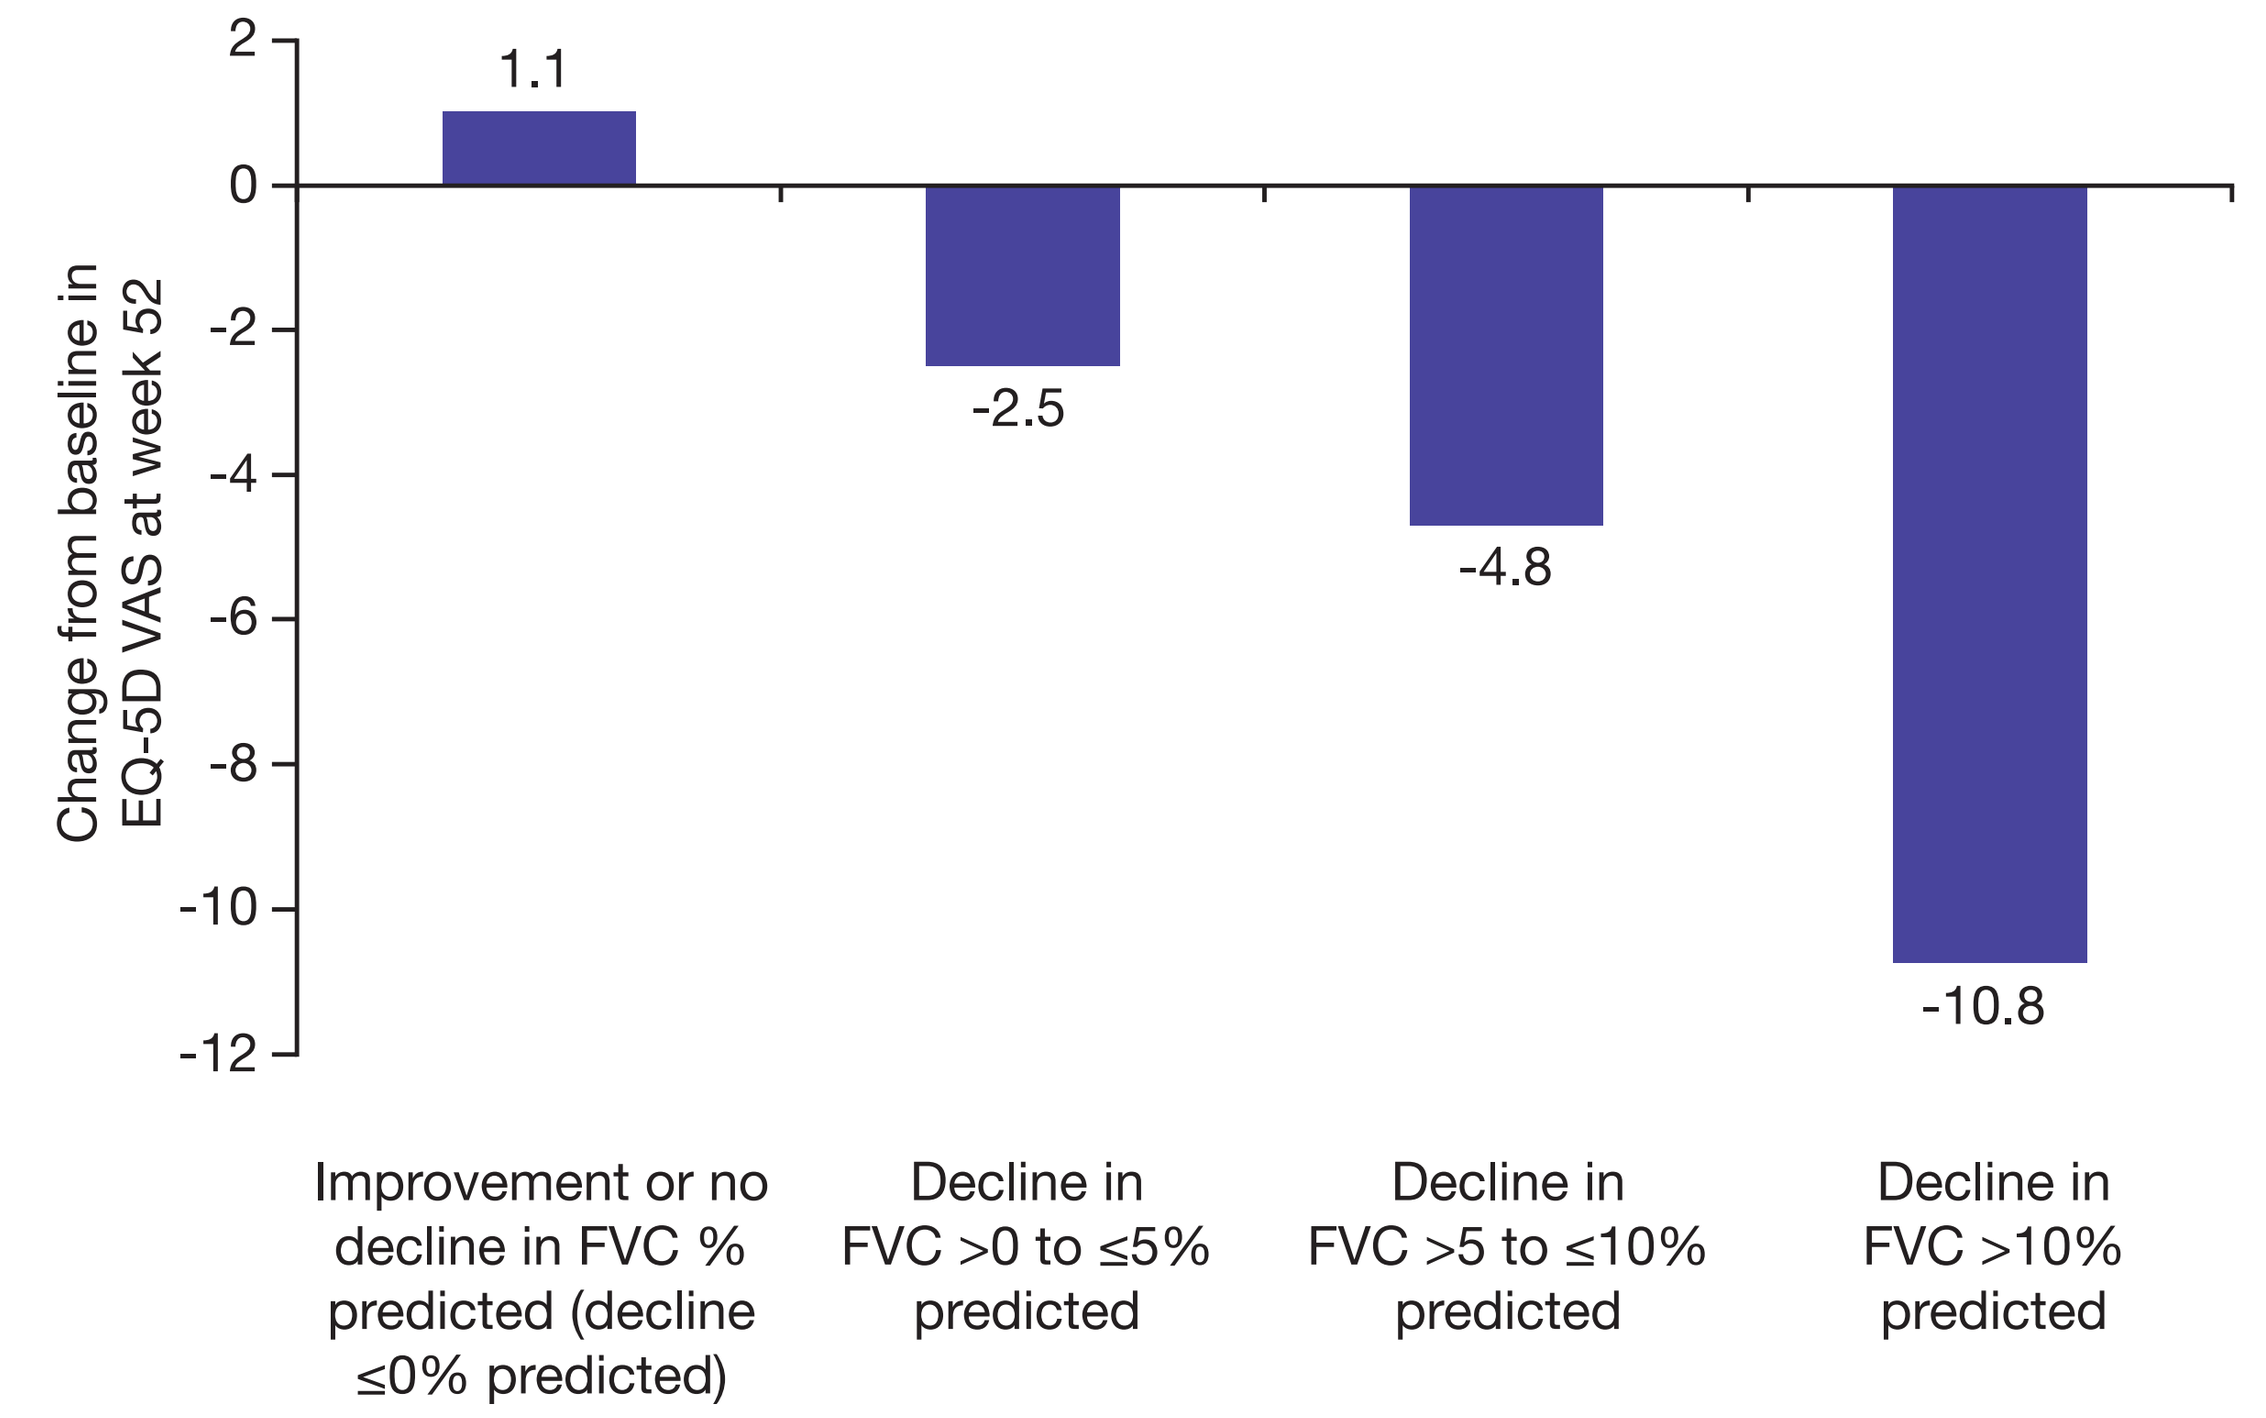

Change from baseline in EQ-5D VAS at week 52
2
0
-2
-4
-6
-8
-10
-12
1.1
-2.5
-4.8
-10.8
Improvement or no decline in FVC % predicted (decline ≤0% predicted)
Decline in FVC >0 to ≤5% predicted
Decline in FVC >5 to ≤10% predicted
Decline in FVC >10% predicted

Conclusions

- **In the INPULSIS® trials, patients with greater declines in FVC % predicted at week 52 had greater worsening in HRQL according to changes in the SGRQ, UCSD-SOBQ, CASA-Q and EQ-VAS.**
- **Additional research is needed to identify specific patient or clinical characteristics that drive decline in HRQL in patients with IPF.**

References

1. Raghu G et al. Am J Respir Crit Care Med 2011; 183:788–824.
2. Belkin A, Swigris JJ. Curr Opin Pulm Med 2013;19:474–9.
3. Richeldi L et al. N Engl J Med 2014;370:2071–82.
4. Kreuter M et al. Respir Res 2017;18:139.
5. Glaspole IN et al. Respirology 2017;22:950–6.
6. Jones PW et al. Respir Med 1991;85 Suppl B:25–31; discussion 33–7.
7. Eakin EG et al. Chest 1998;113:619–24.
8. Crawford B et al. Respir Med 2008;102:1545–55.
9. Rabin R, de Charro F. Ann Med 2001;33:337–43.

Disclosures

The INPULSIS® trials were funded by Boehringer Ingelheim. Medical writing assistance, supported financially by Boehringer Ingelheim, was provided by Julie Fleming and Wendy Morris of FleishmanHillard Fishburn during preparation of this poster. The authors were fully responsible for all content and editorial decisions, were involved at all stages of poster development and have approved the final version. Professor Michael Kreuter has served as a consultant for Boehringer Ingelheim. Nils Schoof is an employee of Boehringer Ingelheim.

Boehringer Ingelheim

Presenter contact information: Professor Michael Kreuter, Center for Interstitial and Rare Lung Diseases, Pneumology, Thoraxklinik, University of Heidelberg, Germany; email: kreuter@uni-heidelberg.de

Poster presented at the American Thoracic Society International Conference, San Diego, CA, USA, 18–23 May 2018.