Safety and tolerability of nintedanib in patients with idiopathic pulmonary fibrosis (IPF): pooled data from six clinical trials

Lisa Lancaster,[1] Paul Hernandez,[2] Yoshikazu Inoue,[3] Daniel Wachtlin,[4] Lazaro Loaiza,[5] Craig S Conoscenti,[6] Manuel Quaresma,[5] Susanne Stowasser,[5] Luca Richeldi[7]

[1]Vanderbilt University Medical Center, Nashville, Tennessee, USA; [2]QEII Health Sciences Centre, Halifax, Nova Scotia, Canada; [3]Clinical Research Center, National Hospital Organization Kinki-Chuo Chest Medical Center, Osaka, Japan; [4]Boehringer Ingelheim Pharma GmbH & Co. KG, Ingelheim am Rhein, Germany; [5]Boehringer Ingelheim International GmbH, Ingelheim am Rhein, Germany; [6]Boehringer Ingelheim Pharmaceuticals, Inc., Ridgefield, Connecticut, USA; [7]Università Cattolica del Sacro Cuore, Fondazione Policlinico A. Gemelli, Rome, Italy.

Introduction

- Nintedanib, an intracellular inhibitor of tyrosine kinases, is an approved treatment for IPF that slows disease progression by reducing decline in forced vital capacity (FVC).[1–5]
- Pooled data from the Phase II TOMORROW and Phase III INPULSIS trials suggested that nintedanib was associated with a 30% reduction in the risk of all-cause mortality compared with placebo over 52 weeks (hazard ratio 0.70 [95% CI: 0.46, 1.08]; p=0.095).[6]
- Nintedanib has an adverse event profile that is manageable for most patients.[1–6]

Aim

- To characterize the safety and tolerability of nintedanib, and investigate predicted life expectancy of patients with IPF treated with nintedanib, based on pooled data from six clinical trials.

Methods

- Data from patients treated with ≥1 dose of nintedanib 150 mg bid in the following trials were pooled:
 - Phase II, 52-week, placebo-controlled TOMORROW trial[1] and its open-label extension[2]
 - Phase III, 52-week, placebo-controlled INPULSIS trials[3] and their open-label extension INPULSIS-ON[4]
 - Phase IIIb trial with a placebo-controlled period of at least 6 months followed by an open-label period of up to 12 months.[5]
- In all these trials, dose reductions to 100 mg bid and treatment interruptions were allowed to manage adverse events.
- All adverse events reported by investigators, irrespective of causality, were analyzed. Adverse events were coded according to the Medical Dictionary for Regulatory Activities (MedDRA) version 19.1.
- Based on the mechanism of action of nintedanib, bleeding, major adverse cardiovascular events (MACE), myocardial infarction and hepatic enzyme elevations were considered of particular interest.
- Event rates per 100 patient exposure–years were calculated based on adverse events with onset after the first dose and up to 28 days (14 days in the TOMORROW trial) after the last dose of study drug.
- Weibull and exponential parametric survival distributions were fitted to pooled Kaplan–Meier survival data for patients treated with ≥1 dose of nintedanib 150 mg bid and placebo in the trials and extrapolated to estimate life expectancy over the long term. The estimated survival of patients treated with nintedanib was compared with that of patients treated with placebo.
- Data from INPULSIS-ON and the Phase IIIb trial were based on a snapshot conducted on 15 April 2017. The other trials had been completed by this date, but the MedDRA coding was updated to obtain consistent coding within the pooled population.

Results

Patients

- A total of 1126 patients treated with ≥1 dose of nintedanib 150 mg bid were included in the pooled population (Table 1).
- Mean (SD) exposure to nintedanib in the pooled population was 27.7 (20.5) months. Median (maximum) exposure was 22.5 (93.1) months. Total exposure was 2599 patient–years.

***Table 1.** Baseline characteristics*

	Pooled population treated with nintedanib (n=1126)	INPULSIS trials	
		Nintedanib (n=638)	Placebo (n=423)
Age, years, mean (SD)	66.9 (8.1)	66.6 (8.1)	67.0 (7.9)
Male, n (%)	882 (78.3)	507 (79.5)	334 (79.0)
Race, n (%)			
White	695 (61.7)	360 (56.4)	248 (58.6)
Asian	316 (28.1)	194 (30.4)	128 (30.3)
Black	2 (0.2)	2 (0.3)	0 (0.0)
Missing*	113 (10.0)	82 (12.9)	47 (11.1)
Body mass index, kg/m^2, mean (SD)	27.9 (4.6)	28.1 (4.6)	27.6 (4.6)
Smoking history, n (%)			
Never smoker	313 (27.8)	174 (27.3)	122 (28.8)
Former smoker	768 (68.2)	435 (68.2)	283 (66.9)
Current smoker	45 (4.0)	29 (4.5)	18 (4.3)
FVC, % predicted, mean (SD)	78.7 (18.4)	79.7 (17.6)	79.3 (18.2)
FEV_1/FVC, %, mean (SD)	81.6 (6.4)	81.7 (5.8)	81.7 (6.0)
DLco, % predicted, mean (SD)	48.2 (13.5)†	47.4 (13.5)	47.0 (13.4)

*In France, regulation did not permit the collection of data on race. †Based on data from 821 patients (DLco was not collected at baseline of the open-label extension of TOMORROW or INPULSIS-ON).

Adverse events

- The most frequently reported adverse event in patients treated with nintedanib in the pooled population was diarrhea (Table 2).
- Rates of adverse events reported in the pooled population were generally lower than those reported in patients treated with nintedanib in the INPULSIS trials (Table 2).

***Table 2.** Most frequent adverse events*

Event rates per 100 patient exposure–years	Pooled population treated with nintedanib (n=1126)	INPULSIS	
		Nintedanib (n=638)	Placebo (n=423)
Diarrhea	76.5	112.6	25.6
Nausea	18.0	34.9	7.0
Nasopharyngitis	15.1	19.6	22.0
Bronchitis	14.5	15.5	15.0
Cough	13.2	16.1	16.2
Progression of IPF*	12.9	11.8	17.7
Vomiting	11.2	17.1	2.7
Upper respiratory tract infection	10.1	12.1	13.3

Adverse events with rate >10 per 100 patient exposure–years in the pooled population are shown.
*Corresponds to MedDRA preferred term 'IPF', which included disease worsening and acute exacerbations.

- The event rates of adverse events of particular interest reported in the pooled population were lower than those reported in patients treated with nintedanib in the INPULSIS trials (Table 3).

***Table 3.** Adverse events of particular interest*

Event rates per 100 patient exposure–years	Pooled population treated with nintedanib (n=1126)	INPULSIS	
		Nintedanib (n=638)	Placebo (n=423)
MACE*	3.7	4.4	2.7
Myocardial infarction†	1.1	1.8	0.5
Bleeding‡	9.3	15.8	10.2
Hepatic enzyme elevation§	12.1	22.2	3.4

*Based on fatal adverse events in the MedDRA system organ classes "cardiac disorders" and "vascular disorders"; fatal and nonfatal events in the subordinate standardized MedDRA query (SMQ) "myocardial infarction"; stroke based on selected preferred terms from the subordinate SMQs "hemorrhagic cerebrovascular conditions" and "ischemic cerebrovascular conditions"; and the MedDRA preferred terms "sudden death", "cardiac death" and "sudden cardiac death". †Based on the subordinate SMQ "myocardial infarction [narrow definition]". ‡Based on the SMQ "hemorrhage terms [excluding laboratory terms]". §Based on a list of MedDRA preferred terms.

- The rate of adverse events that led to permanent dose reduction of nintedanib was 12.8 events per 100 patient exposure–years.
- The event rates of adverse events leading to permanent dose reduction in the pooled population were lower than those in nintedanib-treated patients in the INPULSIS trials (Table 4).

***Table 4.** Adverse events leading to permanent dose reduction*

Event rates per 100 patient exposure–years	Pooled population treated with nintedanib (n=1126)	INPULSIS	
		Nintedanib (n=638)	Placebo (n=423)
Diarrhea	7.4	11.4	0.0
Nausea	1.0	1.8	0.0
Vomiting	0.7	1.2	0.0
Weight decreased	0.6	0.7	0.2

Adverse events leading to permanent dose reduction with rate >0.5 per 100 patient exposure–years in the pooled population are shown.

- The rate of adverse events that led to permanent discontinuation of nintedanib was 23.8 events per 100 patient exposure–years.
- Progression of IPF (which corresponded to the MedDRA preferred term 'IPF' and included disease worsening and acute exacerbations) was the most frequent adverse event leading to permanent discontinuation of nintedanib in the pooled population (Table 5).
- The event rates of adverse events leading to permanent discontinuation of nintedanib in the pooled population were generally lower than those in nintedanib-treated patients in the INPULSIS trials (Table 5).

***Table 5.** Adverse events leading to permanent discontinuation*

Event rates per 100 patient exposure–years	Pooled population treated with nintedanib (n=1126)	INPULSIS	
		Nintedanib (n=638)	Placebo (n=423)
Progression of IPF*	4.2	2.2	5.1
Diarrhea	3.6	4.7	0.2
Nausea	1.0	2.2	0.0
Weight decreased	0.8	1.0	0.2
Decreased appetite	0.7	1.5	0.2

Adverse events leading to permanent treatment discontinuation with rate >0.5 per 100 patient exposure–years in the pooled population are shown. *Corresponds to MedDRA term 'IPF', which included disease worsening and acute exacerbations.

Survival

- Based on the Weibull distribution, mean (95% CI) survival was estimated as 11.6 (9.6, 14.1) years in nintedanib-treated patients and 3.7 (2.5, 5.4) years in placebo-treated patients in the pooled population (Figure 1). Median survival was estimated as 8.5 years in nintedanib-treated patients and 3.3 years in placebo-treated patients.

***Figure 1.** Estimated time to death using the Weibull distribution*

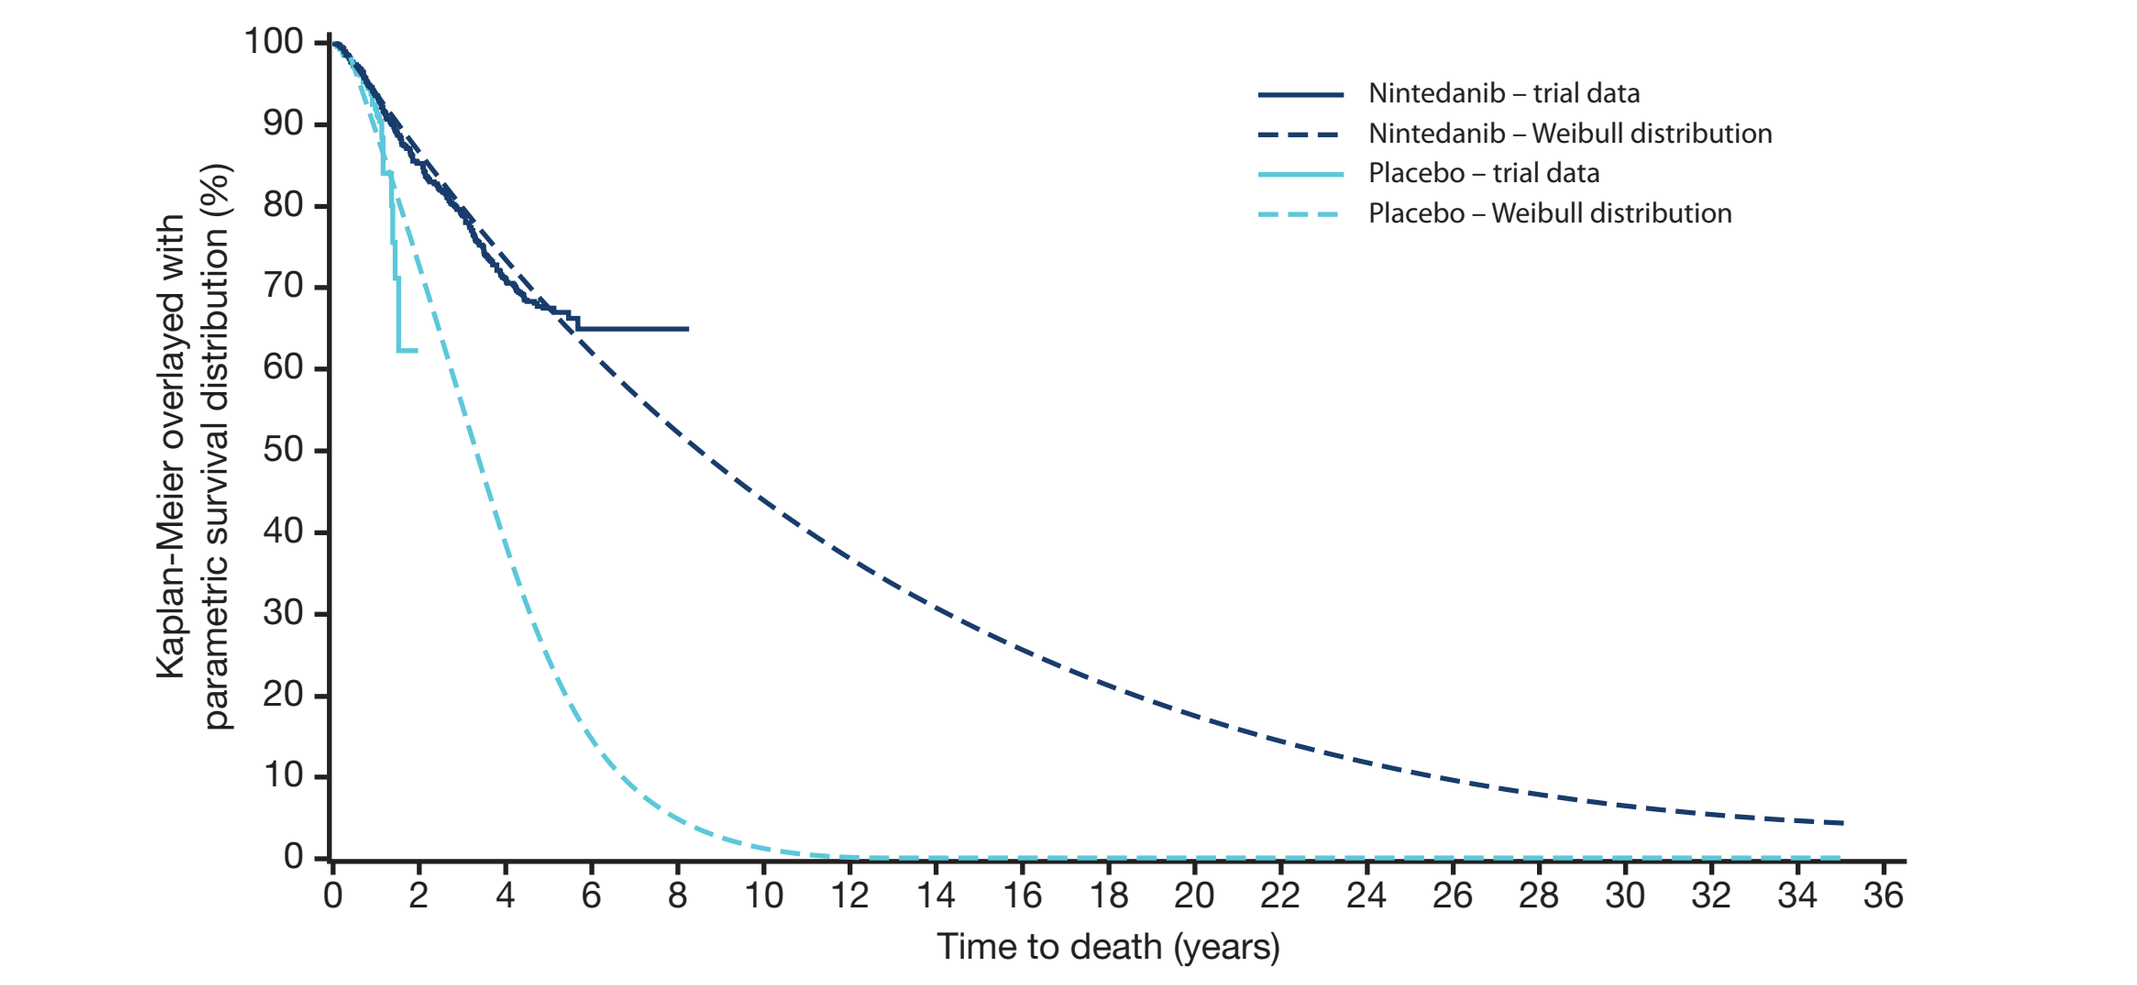

Nintedanib – trial data
Nintedanib – Weibull distribution
Placebo – trial data
Placebo – Weibull distribution
Kaplan-Meier overlayed with parametric survival distribution (%)
Time to death (years)

- Based on exponential distribution, mean (95% CI) survival was estimated as 13.1 (11.6, 14.9) years in nintedanib-treated patients and 10.1 (7.8, 13.0) years in placebo-treated patients in the pooled population (Figure 2). Median survival was estimated as 9.1 years in nintedanib-treated patients and 7.0 years in placebo-treated patients.
- Model fit based on Akaike's Information Criterion (AIC) was superior (lower) for the Weibull model compared with the exponential model, especially for placebo-treated patients (428 versus 440).

***Figure 2.** Estimated time to death using exponential distribution*

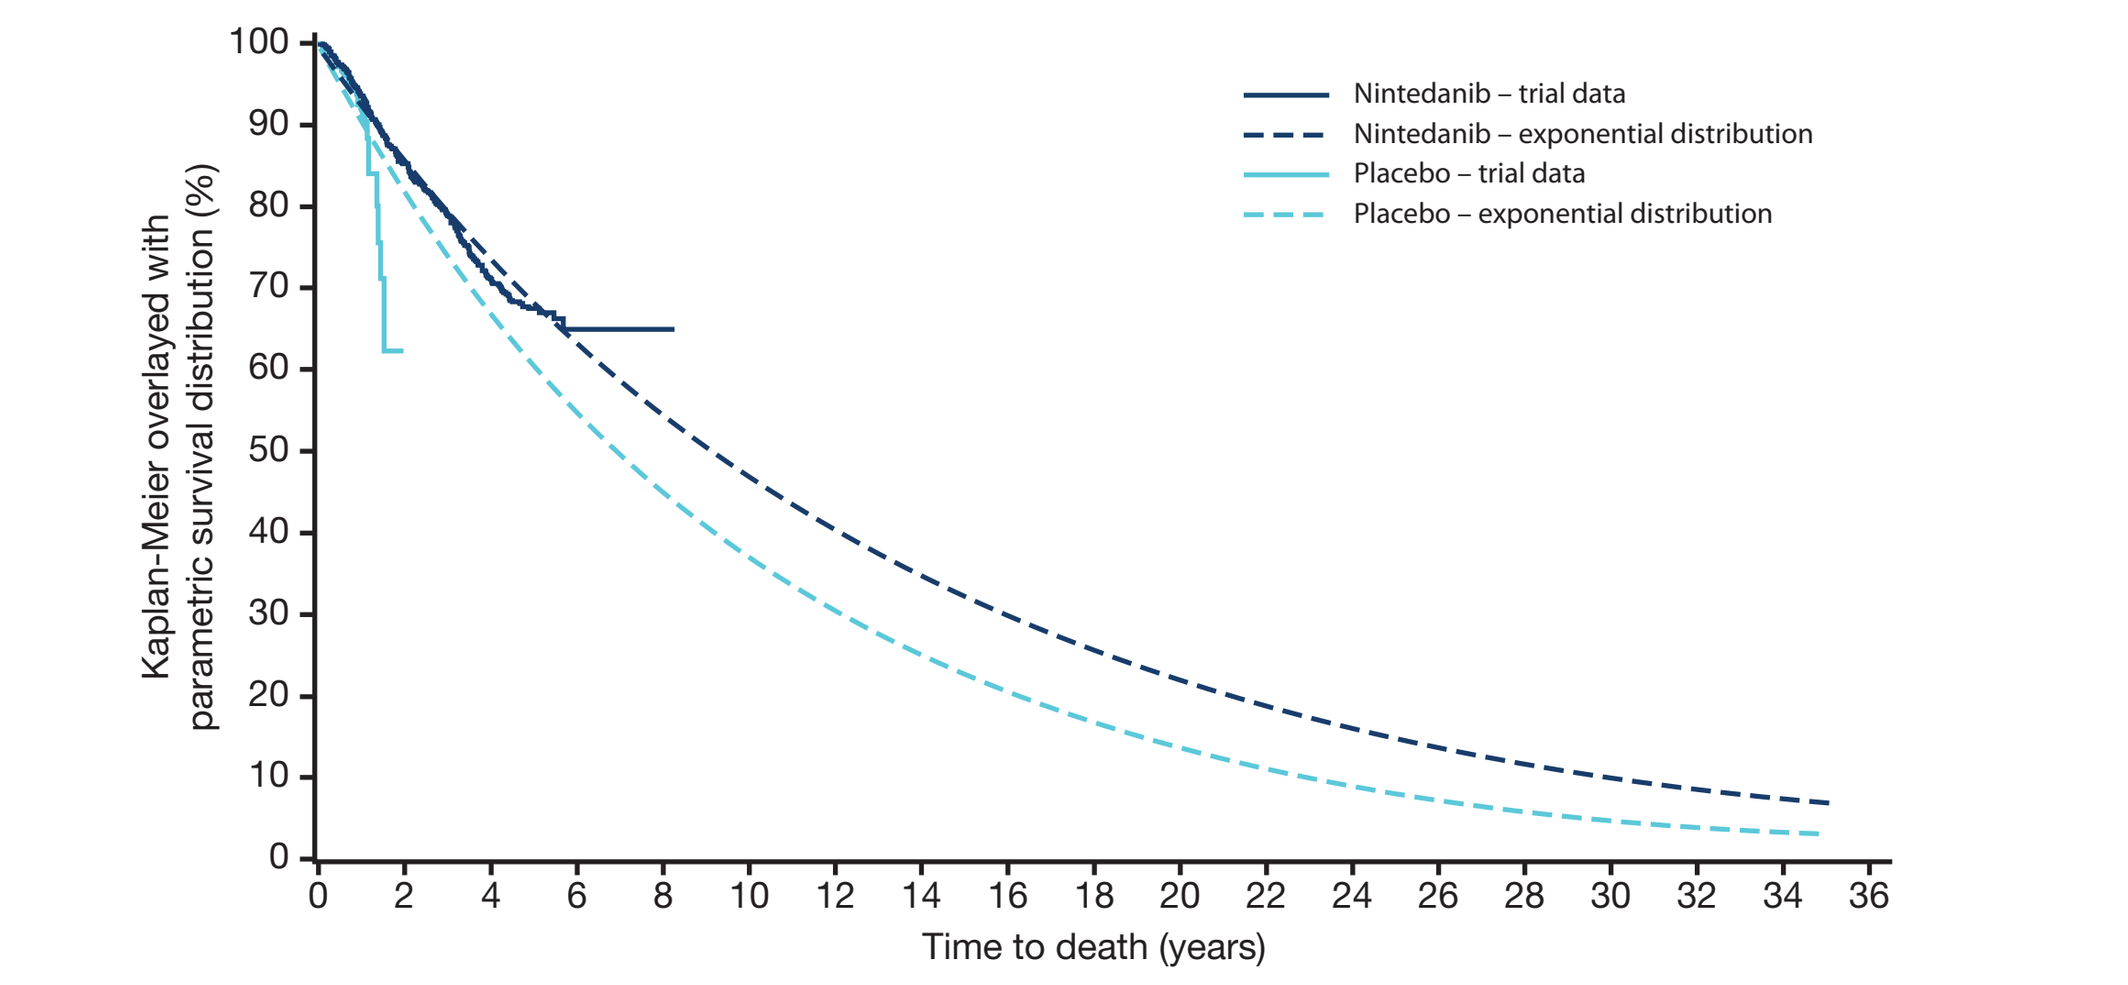

Nintedanib – trial data
Nintedanib – exponential distribution
Placebo – trial data
Placebo – exponential distribution
Kaplan-Meier overlayed with parametric survival distribution (%)
Time to death (years)

Key limitations and caveats

- There was selection bias in the nintedanib-treated patients who entered the open-label extension trials, in that these patients were likely better able to tolerate nintedanib than the nintedanib-treated patients who dropped out during the parent trial.
- Estimates of life expectancy obtained from extrapolation rather than observed values should be interpreted with caution.

Conclusions

- **In 1126 patients with IPF treated with nintedanib for a mean duration of 28 months and up to a maximum of 93 months, adverse events were manageable for most patients.**
- **The adverse event profile of nintedanib in the pooled population was consistent with that observed in the INPULSIS trials. No new safety signals were observed.**
- **Exploratory analyses suggest that patients treated with nintedanib have longer life expectancy than patients who do not receive treatment.**

References

1. Richeldi L et al. N Engl J Med 2011;365:1079–87.
2. Richeldi L et al. Thorax 2017; pii: thoraxjnl-2016-209701; Epub ahead of print.
3. Richeldi L et al. N Engl J Med 2014;370:2071–82.
4. Crestani B et al. Eur Respir J 2017;50 (suppl 61):OA3402.
5. Lancaster L et al. Poster presented at the American Thoracic Society International Conference, San Diego, USA, 18–23 May 2018 (poster number: 215).
6. Richeldi L et al. Respir Med 2016;113:74–9.

Disclosures

These trials were funded by Boehringer Ingelheim. Medical writing assistance, supported financially by Boehringer Ingelheim, was provided by Julie Fleming and Wendy Morris of FleishmanHillard Fishburn, London, UK during the preparation of this poster. The authors were fully responsible for all content and editorial decisions, were involved at all stages of poster development and have approved the final version. Dr Lisa Lancaster has served on advisory boards for Genentech, Global Blood Therapeutics, Boehringer Ingelheim, Veracyte and Bellopheron, has provided disease state education for Genentech and Boehringer Ingelheim, and has served as a principal investigator in clinical trials in IPF for Genentech, Global Blood Therapeutics, Celgene, FibroGen and Boehringer Ingelheim. Professor Luca Richeldi reports personal fees from Boehringer Ingelheim, Biogen, Cipla, ImmuneWorks, MedImmune, Pliant Therapeutics, Roche, Sanofi-Aventis, Shionogi, and Takeda.

Presenter contact information: Dr Lisa Lancaster, Vanderbilt University Medical Center, Nashville, Tennessee, USA; lisa.lancaster@vanderbilt.edu

Poster presented at the American Thoracic Society International Conference, San Diego, CA, USA, 18–23 May 2018.